Comprehensive Kinase Profile of Pacritinib, a Non-Myelosuppressive JAK2 Kinase Inhibitor in Phase 3 Development in Primary and Post-ET/PV Myelofibrosis

Jack W. Singer, M.D.[1], Suliman Al-Fayoumi, Ph.D., M.B.A.[1*], Haiching Ma[2*], Rami S. Komrokji, M.D.[3], Ruben Mesa, M.D.[4] and Srdan Verstovsek, M.D., Ph.D.[5]

[1]CTI BioPharma Corp, Seattle, WA; [2]Reaction Biology, Malvern, PA; [3]Department of Malignant Hematology, H. Lee Moffitt Cancer Center & Research Institute, Tampa, FL; [4]Division of Hematology and Medical Oncology, Mayo Clinic Cancer Center, Scottsdale, AZ; [5]Department of Leukemia, Division of Cancer Medicine, The University of Texas MD Anderson Cancer Center, Houston, TX; *Not a member of ASH

BACKGROUND

Pacritinib, an ATP binding site multikinase inhibitor with low nM activity against JAK2 and JAK2 V617F, is in Phase 3 development in myelofibrosis.

In preclinical models, and in Phase 1 studies in both non-Hodgkin lymphoma and myelofibrosis, it appeared unique among this class of agents for its lack of myelosuppression at doses that inhibit the JAK2/STAT3 pathway and result in clinical efficacy.

In two Phase 2 studies in myelofibrosis that had no exclusion restriction for thrombocytopenia, pacritinib was effective at reducing spleen size and symptoms regardless of starting platelet counts. There was no apparent treatment-related decline in platelet counts regardless of starting levels.

Pacritinib did not cause worsening anemia or cause an increase in RBC transfusion requirements. There was no apparent non-hematological cumulative toxicity and dose adjustments were rarely required (Verstovsek et al.; Blood 122:21, 2013).

To help elucidate the mechanisms for pacritinib's lack of hematopoietic suppression despite its low nM inhibition of JAK2/STAT3, we performed a kinome-wide screen to evaluate its spectrum of kinase inhibition.

KINASE ASSAY METHODS

In vitro profiling of the 429 member kinase panel was performed at Reaction Biology Corporation using the "HotSpot" assay platform (Nature Biotechnology; 29:1039-45, 2011).

For the screening assays, pacritinib was tested at 100 nM. After subtraction of background derived from control reactions containing inactive enzyme, kinase activity data were expressed as the percent remaining kinase activity in test samples compared to DMSO control.

Dose responses from 1-100 nM were performed on all kinases that were inhibited by >50% at 100 nM and curve fits were obtained using Prism (GraphPad Software).

Kinome tree representations were prepared using Kinome Mapper.
http://www.reactionbiology.com/apps/kinome/mapper/LaunchKinome.htm

JAK AND FLT3 KINASES

Kinase	IC50 (nM)	Ligand
JAK1	No inhibition at 100	IL-2, IFN
JAK2	6.0	EPO-R, MPL
JAK2 (V617F)	9.4	
FLT3	14.8	FLT3 ligand
FLT3 (ITD)	*13.4*	
FLT3 (D835Y)	*4.7*	
TYK2	27.0	IL-12, IFN α and β, IL-10, IL-2
JAK3	18.3	Type 1 cytokine family receptors in lymphoid and NK cells; IL-8 in neutrophils

NON-JAK/FLT3 TYROSINE KINASES

Kinase	IC50 (nM)	Ligand
TRKC	18.4	NT-Growth Factor (neuronal tumors)
TNK1	15.0	Non-receptor TK (ras/raf/MAPK pathway)
ROS1	18.4	Insulin receptor family; proto-oncogene; Activating fusions (NSCLC, cholangiocarcinoma)
C-KIT	43.8	C-KIT Ligand (Steele Factor)
C-SRC	46.7	Proto-oncogene
C-FMS	39.5	CSF1 receptor; mutations in CMML, AML; Nurse-Like Cells in CLL and MCL

NON TYROSINE KINASES OF INTEREST

Kinase	IC50 (nM)	Ligand
HIPK4	14.5	P53 serine 9
IRAK1	13.6	IL-1 receptor ST kinase (downstream from TLR). Proven target in MDS and CMML; associated with microenvironmental tumor interactions.

PACRITINIB KINOME ACTIVITY MAP

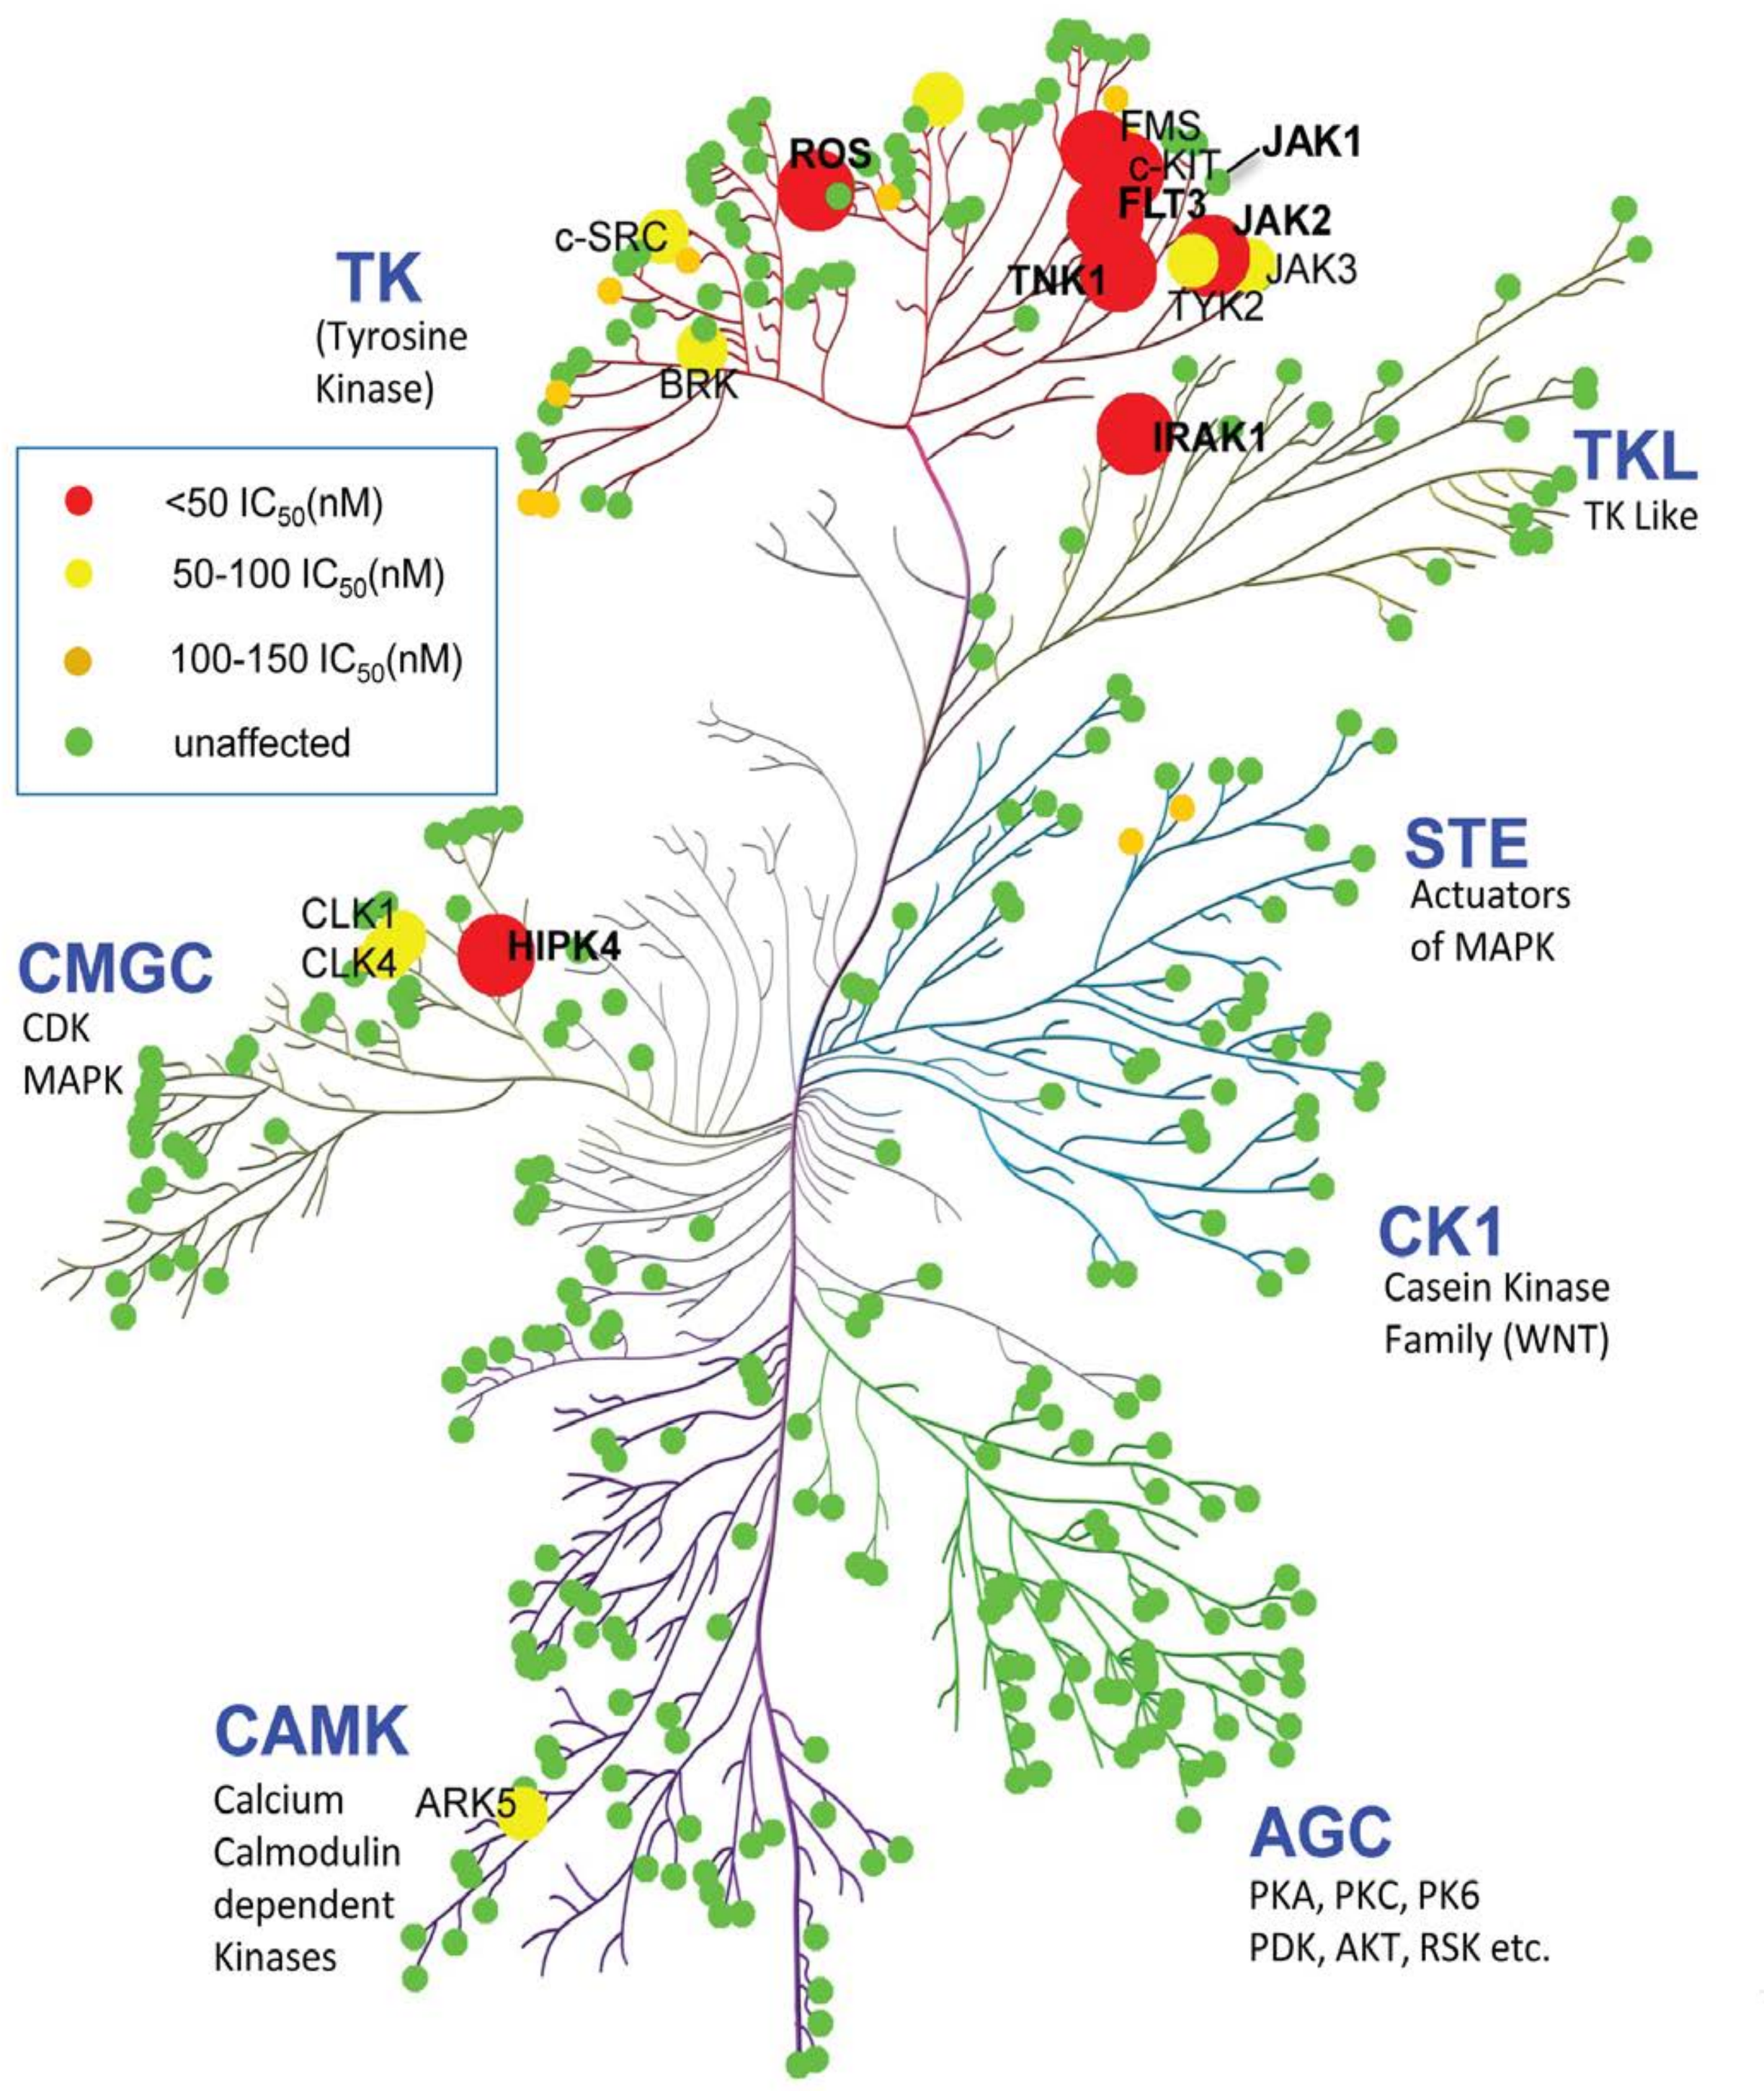
TK
(Tyrosine Kinase)
ROS
c-SRC
BRK
FMS
C-KIT
FLT3
JAK1
JAK2
JAK3
TNK1
TYK2
IRAK1
TKL
TK Like
<50 IC50(nM)
50-100 IC50(nM)
100-150 IC50(nM)
unaffected
STE
Actuators of MAPK
CMGC
CDK
MAPK
CLK1
CLK4
HIPK4
CK1
Casein Kinase Family (WNT)
CAMK
Calcium Calmodulin dependent Kinases
ARK5
AGC
PKA, PKC, PK6
PDK, AKT, RSK etc.

HUMAN PK – CMAX FOR PROTEIN BOUND AND FREE PACRITINIB

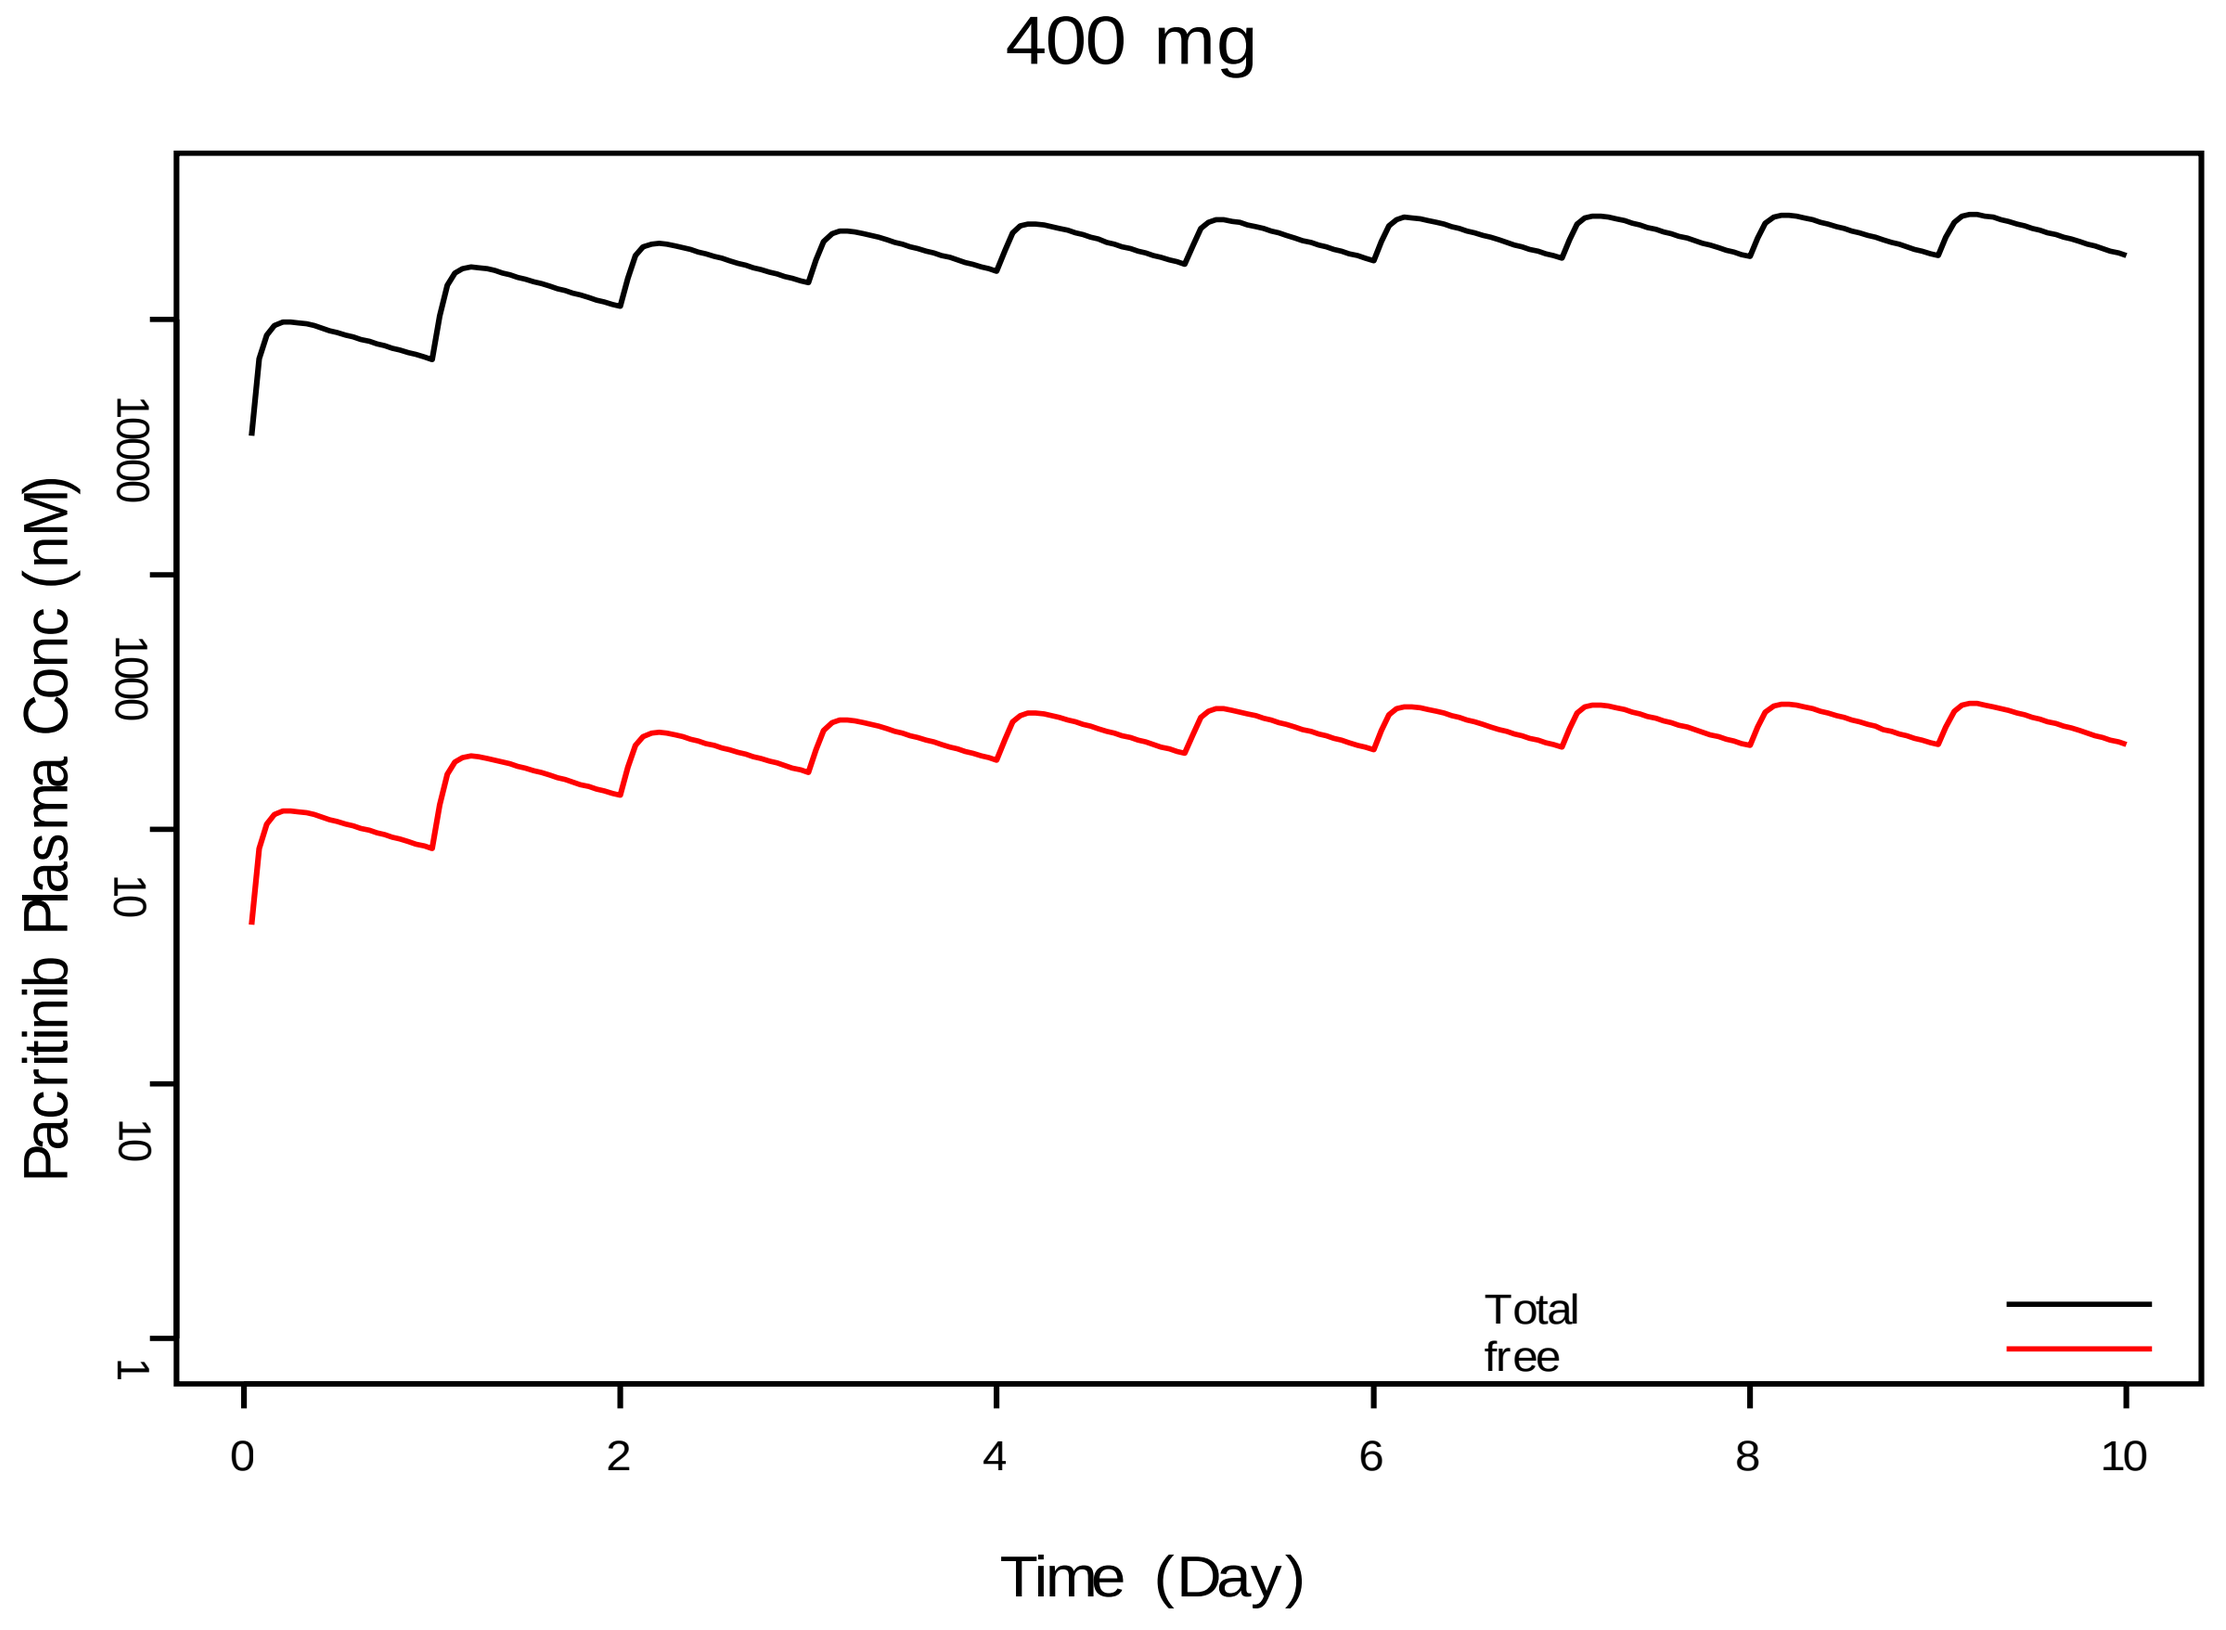
400 mg
Pacritinib Plasma Conc (nM)
Total
free
0
2
4
6
8
10
Time (Day)

DISCUSSION & CONCLUSIONS

- Pacritinib is an oral multikinase inhibitor with specificity for kinases in several unrelated families.
- Steady-state concentrations of up to 200 nM of free pacritinib are achieved with standard dosing and acceptable side effects.
- Pacritinib suppresses all members of the JAK/FLT pathways, with the exception of JAK1 at low nM concentrations. It is inactive against JAK1 at 100 nM.
- The inhibition of IRAK1 and c-fms (CSF1R) may contribute to its anti-inflammatory activity and early onset of symptomatic benefit in myelofibrosis.
- Although pacritinib suppresses the JAK2/STAT3 pathway, pacritinib does not cause myelosuppression. We hypothesize that this may be due in part to its ability to decrease production of negative regulators of hematopoiesis through suppression of c-fms and IRAK1.
- The kinase profile of pacritinib suggests its potential therapeutic utility in AML, MDS, and particularly in CMML due to its potent inhibition of c-fms, IRAK1, JAK2 and FLT3. These potential indications are under exploration in ongoing clinical trials.
- The potential for pacritinib to interfere with microenvironmental-tumor interactions through suppression of IRAK1 and c-fms, along with its lack of myelosuppression, suggest its potential for use in combination with established therapies in a variety of liquid and solid tumors.

Abstract #1874 - ASH 2014